Alzheimer's Disease Preclinical Efficacy Database (AlzPED): Improving the Scientific Rigor and Reproducibility of Preclinical Research in Alzheimer's Disease

Shreaya Chakroborty, Jaya Viswanathan, Maria Fe Lanfranco Gallofre, Zane Martin, Jean Yuan, Suzana Petanceska, and Lorenzo Refolo

National Institute on Aging, Bethesda, MD, USA

BACKGROUND

A major challenge to the successful development of therapies for Alzheimer's disease (AD) is the poor translation of preclinical efficacy from animal models to the clinic. Key contributing factors to the unsuccessful translation of therapeutic efficacy include:

- the failure of animal models to fully recapitulate human AD,
- poor rigor in study design, methodology and data analysis,
- failure to match outcome measures used in preclinical animal studies and clinical studies,
- poor reproducibility of published data, and
- publication bias in favor of reporting positive findings and under reporting negative findings.

To address key factors contributing to poor translation of preclinical efficacy from animal models to the clinic in AD therapy development, several advisory meetings and workshops including the National Institutes of Health (NIH) AD Summits in 2012 and 2015 were held. In response to expert recommendations from these meetings, the National Institute on Aging (NIA) and the NIH Library have created an open science knowledge portal – the **Alzheimer's Disease Preclinical Efficacy Database** or AlzPED. Through the following capabilities, AlzPED is intended to guide the development and implementation of strategies and recommendations for standardized best practices for the rigorous preclinical testing of AD candidate therapeutics:

1. Publicly available database of preclinical efficacy studies that houses experimental designs and analyses of **positive and negative data** to overcome publication bias.

2. Knowledge platform for data sharing, mining and analysis of experimental details, designs, data and methods relating to the preclinical testing of candidate therapeutic agents in AD animal models.

3. Database identifying critical experimental design elements and methodology missing from studies, making them susceptible to misinterpretation and reducing their reproducibility and translational value.

CAPABILITIES AND SCOPE

AlzPED has the following capabilities:

- Provides researchers and information scientists with a facile way to survey existing AD preclinical therapy development literature and raise awareness about the **elements of rigorous study design** and **requirements for transparent reporting**.
- Currently hosts curated summaries from **1298** preclinical efficacy studies published between 2000 and 2021.
- Influences the development and implementation of reproducibility strategies including guidelines for standardized best practices for the rigorous preclinical testing of AD candidate therapeutics.
- Provides search capability across relevant translational criteria data sets and external databases:
 - Therapy Type (16 therapy types)
 - Therapeutic Agent (1123 agents)
 - Therapeutic Target (251 targets)
 - Animal Model (210 models)
 - Principal Investigator
 - Funding Source
 - Related Publications (PubMed)
 - Therapeutic Agents (PubChem and DrugBank)
 - Therapeutic Targets (Open Targets and Pharos)
 - Animal Model (Alzforum)
 - Related Clinical Trials (ClinicalTrials.gov)
 - Related Patents (Google Patents and USTPO)
- Provides funding agencies with a tool for enforcement of requirements for transparent reporting and rigorous study design.
- Provides a platform for creating citable reports/preprints of **unpublished studies**, including studies with **negative data**.
- **Reports on the rigor of each study by summarizing the elements of experimental design.**

RIGOR REPORT CARD & REPORTING OF STANDARDIZED SET OF DESIGN ELEMENTS

Experimental Design *Rigor Report Card*

Is the following information reported in the study?:

- ✓ Power/Sample Size Calculation
- ✓ Blinded for Treatment
- ✗ Pharmacokinetic Measures
- ✗ Toxicology Measures
- ✗ Biomarkers
- ✓ Formulation
- ✓ Duration of Treatment
- ✓ Age of Animal at the Beginning of Treatment
- ✓ Sex as a Biological Variable
- ✗ Number of Premature Deaths
- ✓ Statistical Plan
- ✓ Inclusion/Exclusion Criteria Included
- ✓ Randomized into Groups
- ✓ Blinded for Outcome Measures
- ✗ Pharmacodynamic Measures
- ✗ ADME Measures
- ✓ Dose
- ✓ Route of Delivery
- ✓ Frequency of Administration
- ✓ Age of Animal at the End of Treatment
- ✓ Study Balanced for Sex as a Biological Variable
- ✓ Number of Excluded Animals
- ✓ Genetic Background
- ✓ Conflict of Interest

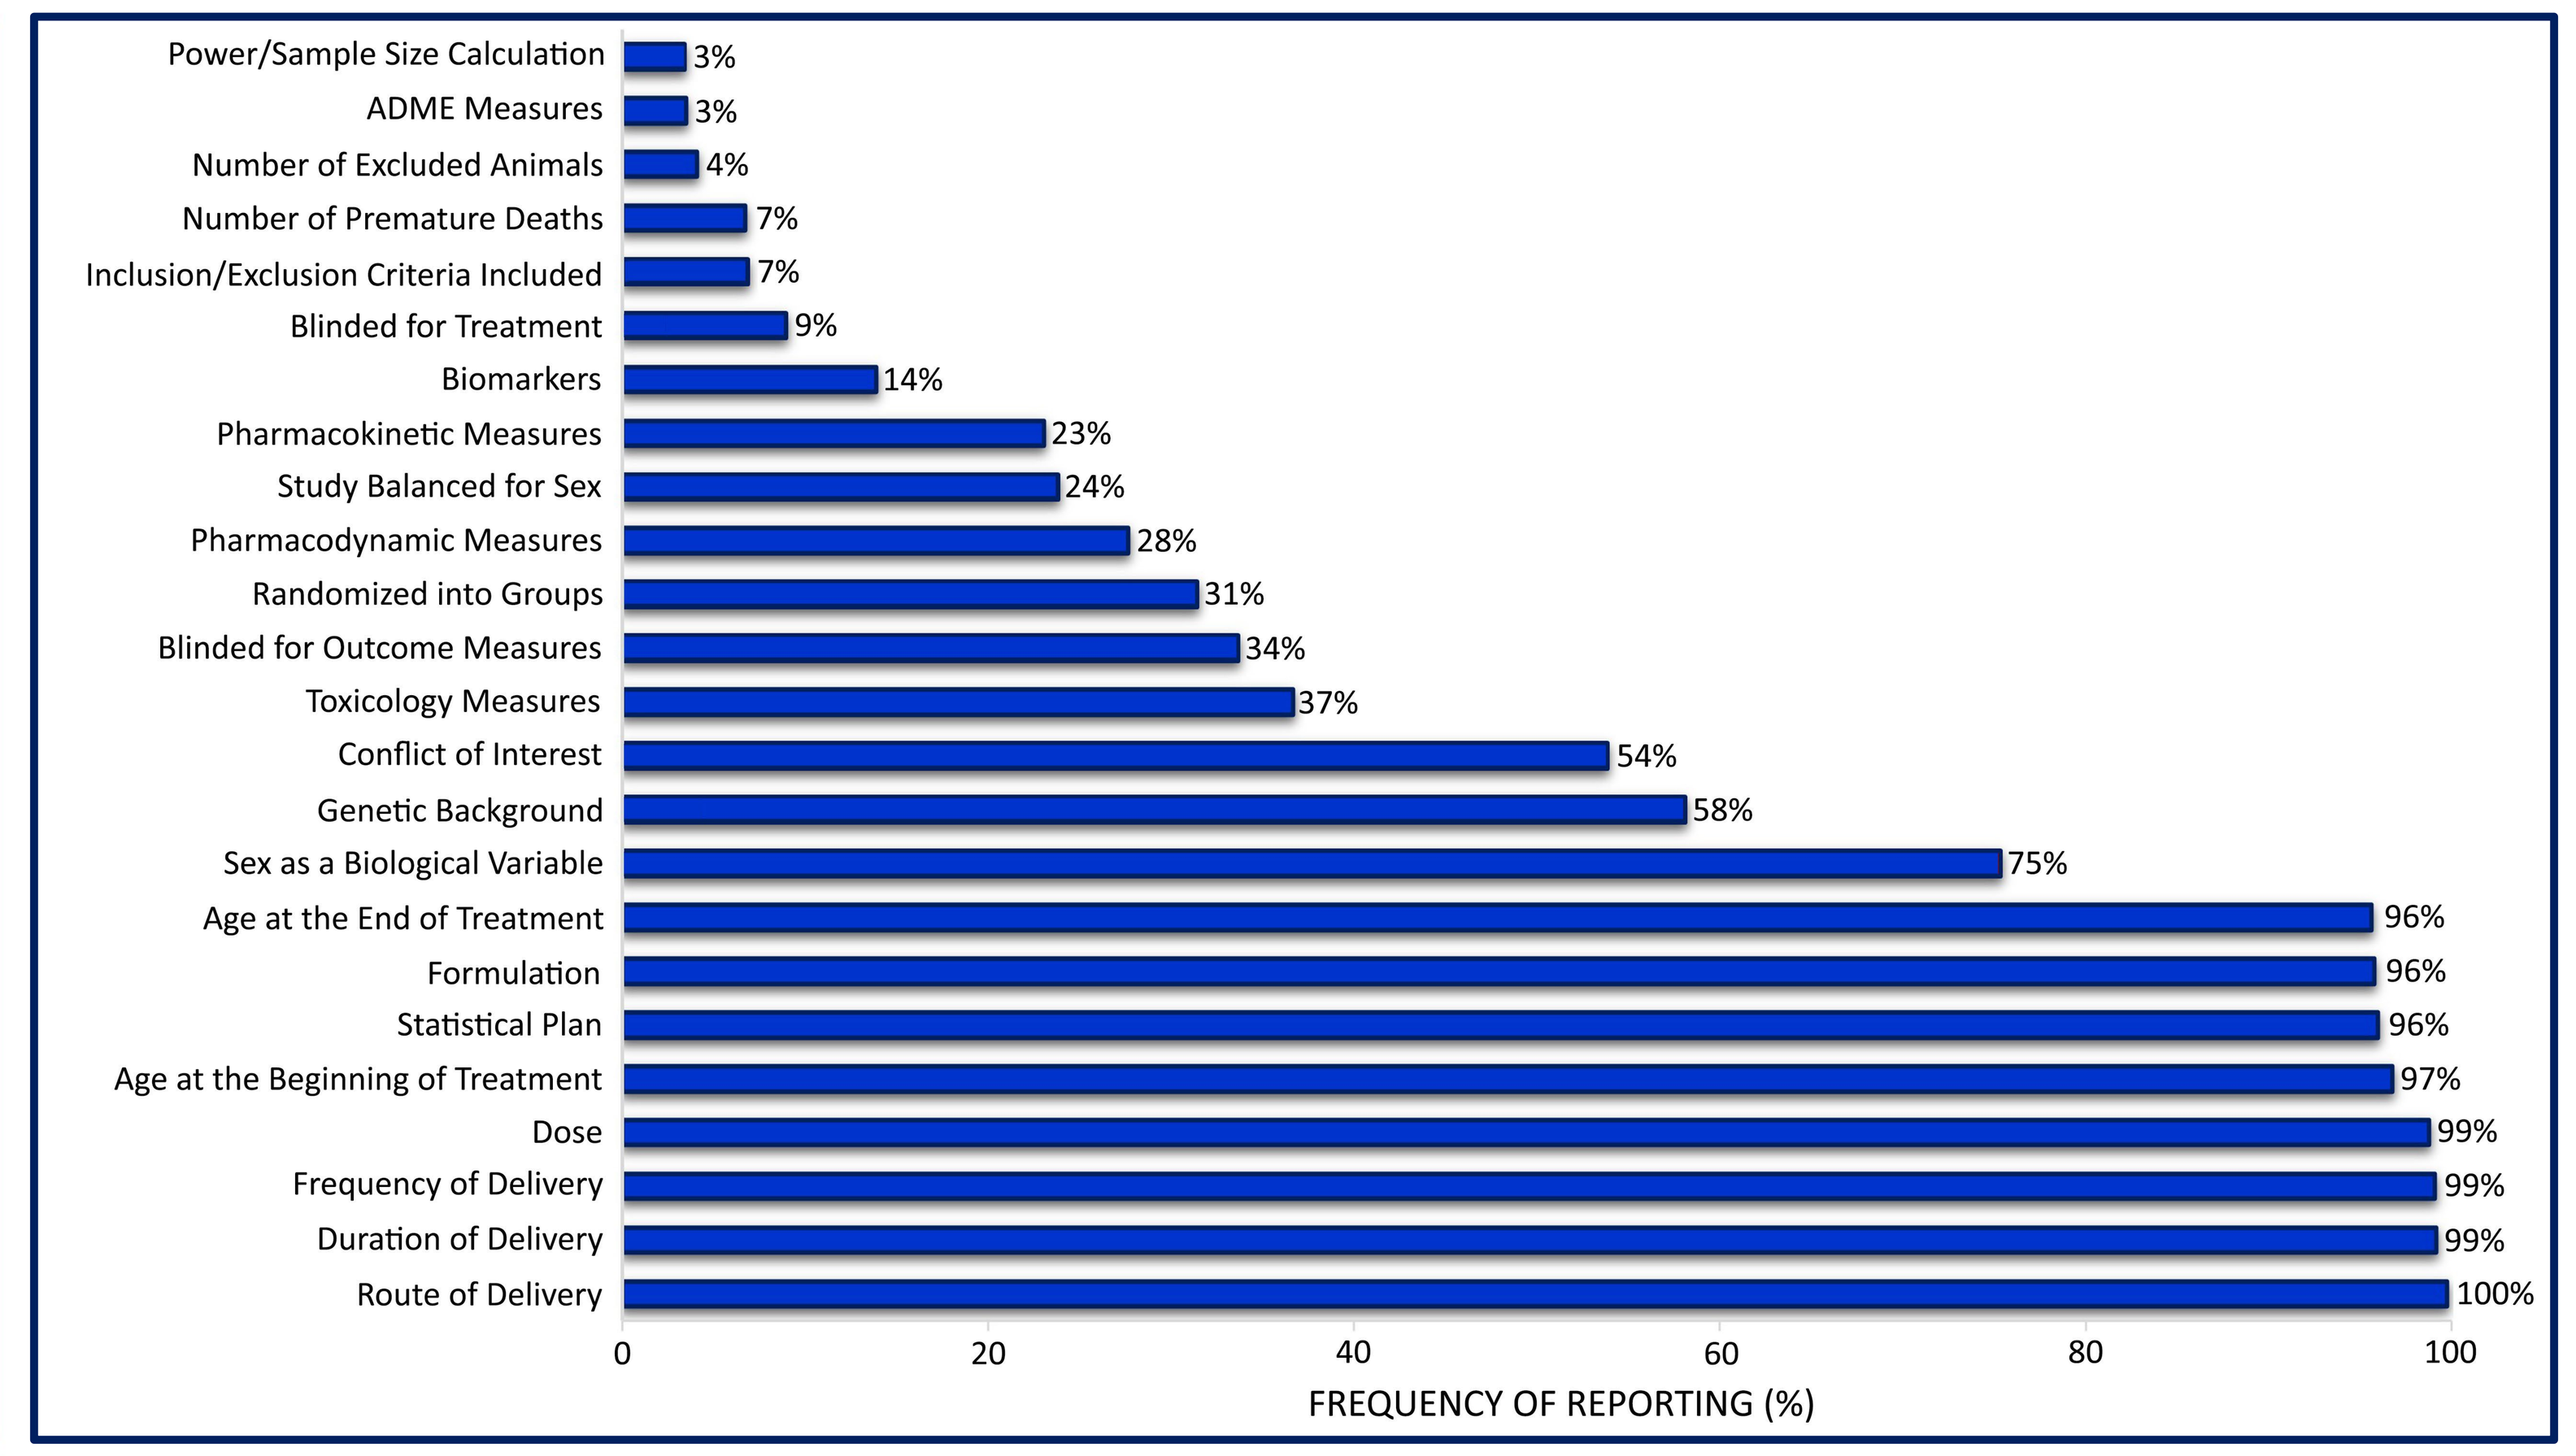

Power/Sample Size Calculation 3%
ADME Measures 3%
Number of Excluded Animals 4%
Number of Premature Deaths 7%
Inclusion/Exclusion Criteria Included 7%
Blinded for Treatment 9%
Biomarkers 14%
Pharmacokinetic Measures 23%
Study Balanced for Sex 24%
Pharmacodynamic Measures 28%
Randomized into Groups 31%
Blinded for Outcome Measures 34%
Toxicology Measures 37%
Conflict of Interest 54%
Genetic Background 58%
Sex as a Biological Variable 75%
Age at the End of Treatment 96%
Formulation 96%
Statistical Plan 96%
Age at the Beginning of Treatment 97%
Dose 99%
Frequency of Delivery 99%
Duration of Delivery 99%
Route of Delivery 100%
0 20 40 60 80 100
FREQUENCY OF REPORTING (%)

Left: AlzPED is designed to monitor the scientific rigor of curated studies with a **"Rigor Report Card"** consisting of a standardized set of 24 experimental design elements recommended for preclinical efficacy studies. The Rigor Report Card demonstrates which design elements are reported in the curated study, and which elements are not, thereby providing a report on rigor of the study and identifying critical elements of experimental design missing from the study. **Right:** Graph shows the percentage of studies reporting the standardized set of 24 experimental design elements. There is significant under-reporting of critical elements of methodology such as power calculation, blinding, randomization, balancing for sex, these being reported by fewer than 35% of the curated studies. Most of the studies report dose and formulation of the therapeutic agent being tested and treatment paradigms (route, frequency and duration of treatment). Data is presented as percentage reported, calculated from 1298 published preclinical studies curated to AlzPED.

REPORTING TRENDS FOR 9 CORE EXPERIMENTAL DESIGN ELEMENTS

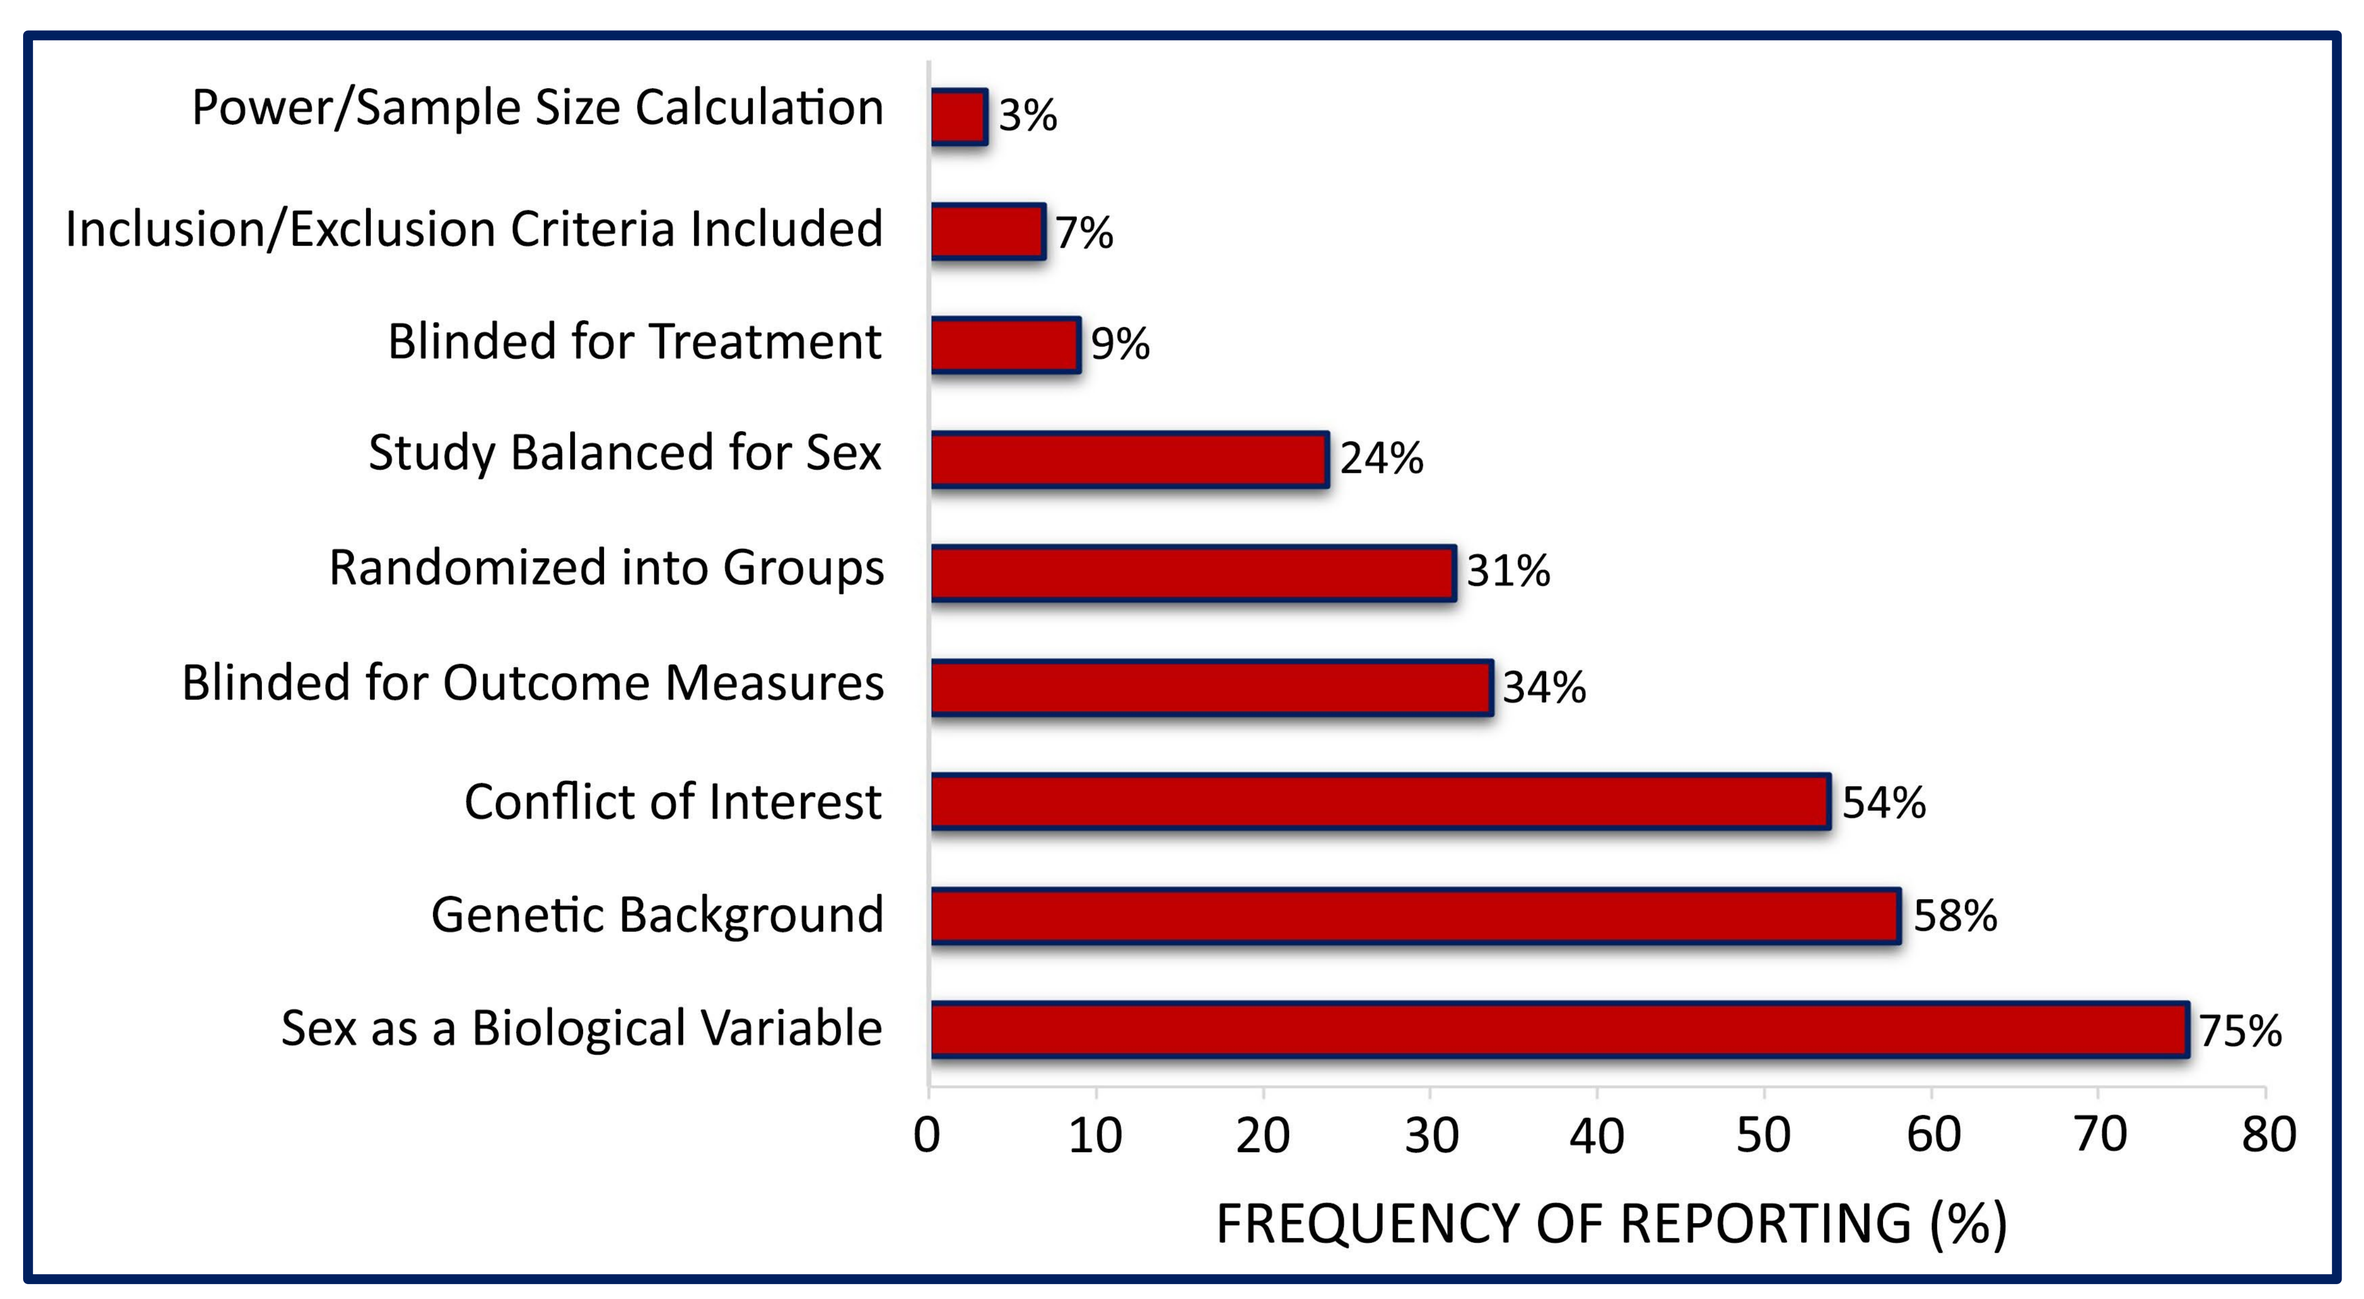

Power/Sample Size Calculation 3%
Inclusion/Exclusion Criteria Included 7%
Blinded for Treatment 9%
Study Balanced for Sex 24%
Randomized into Groups 31%
Blinded for Outcome Measures 34%
Conflict of Interest 54%
Genetic Background 58%
Sex as a Biological Variable 75%
0 10 20 30 40 50 60 70 80
FREQUENCY OF REPORTING (%)

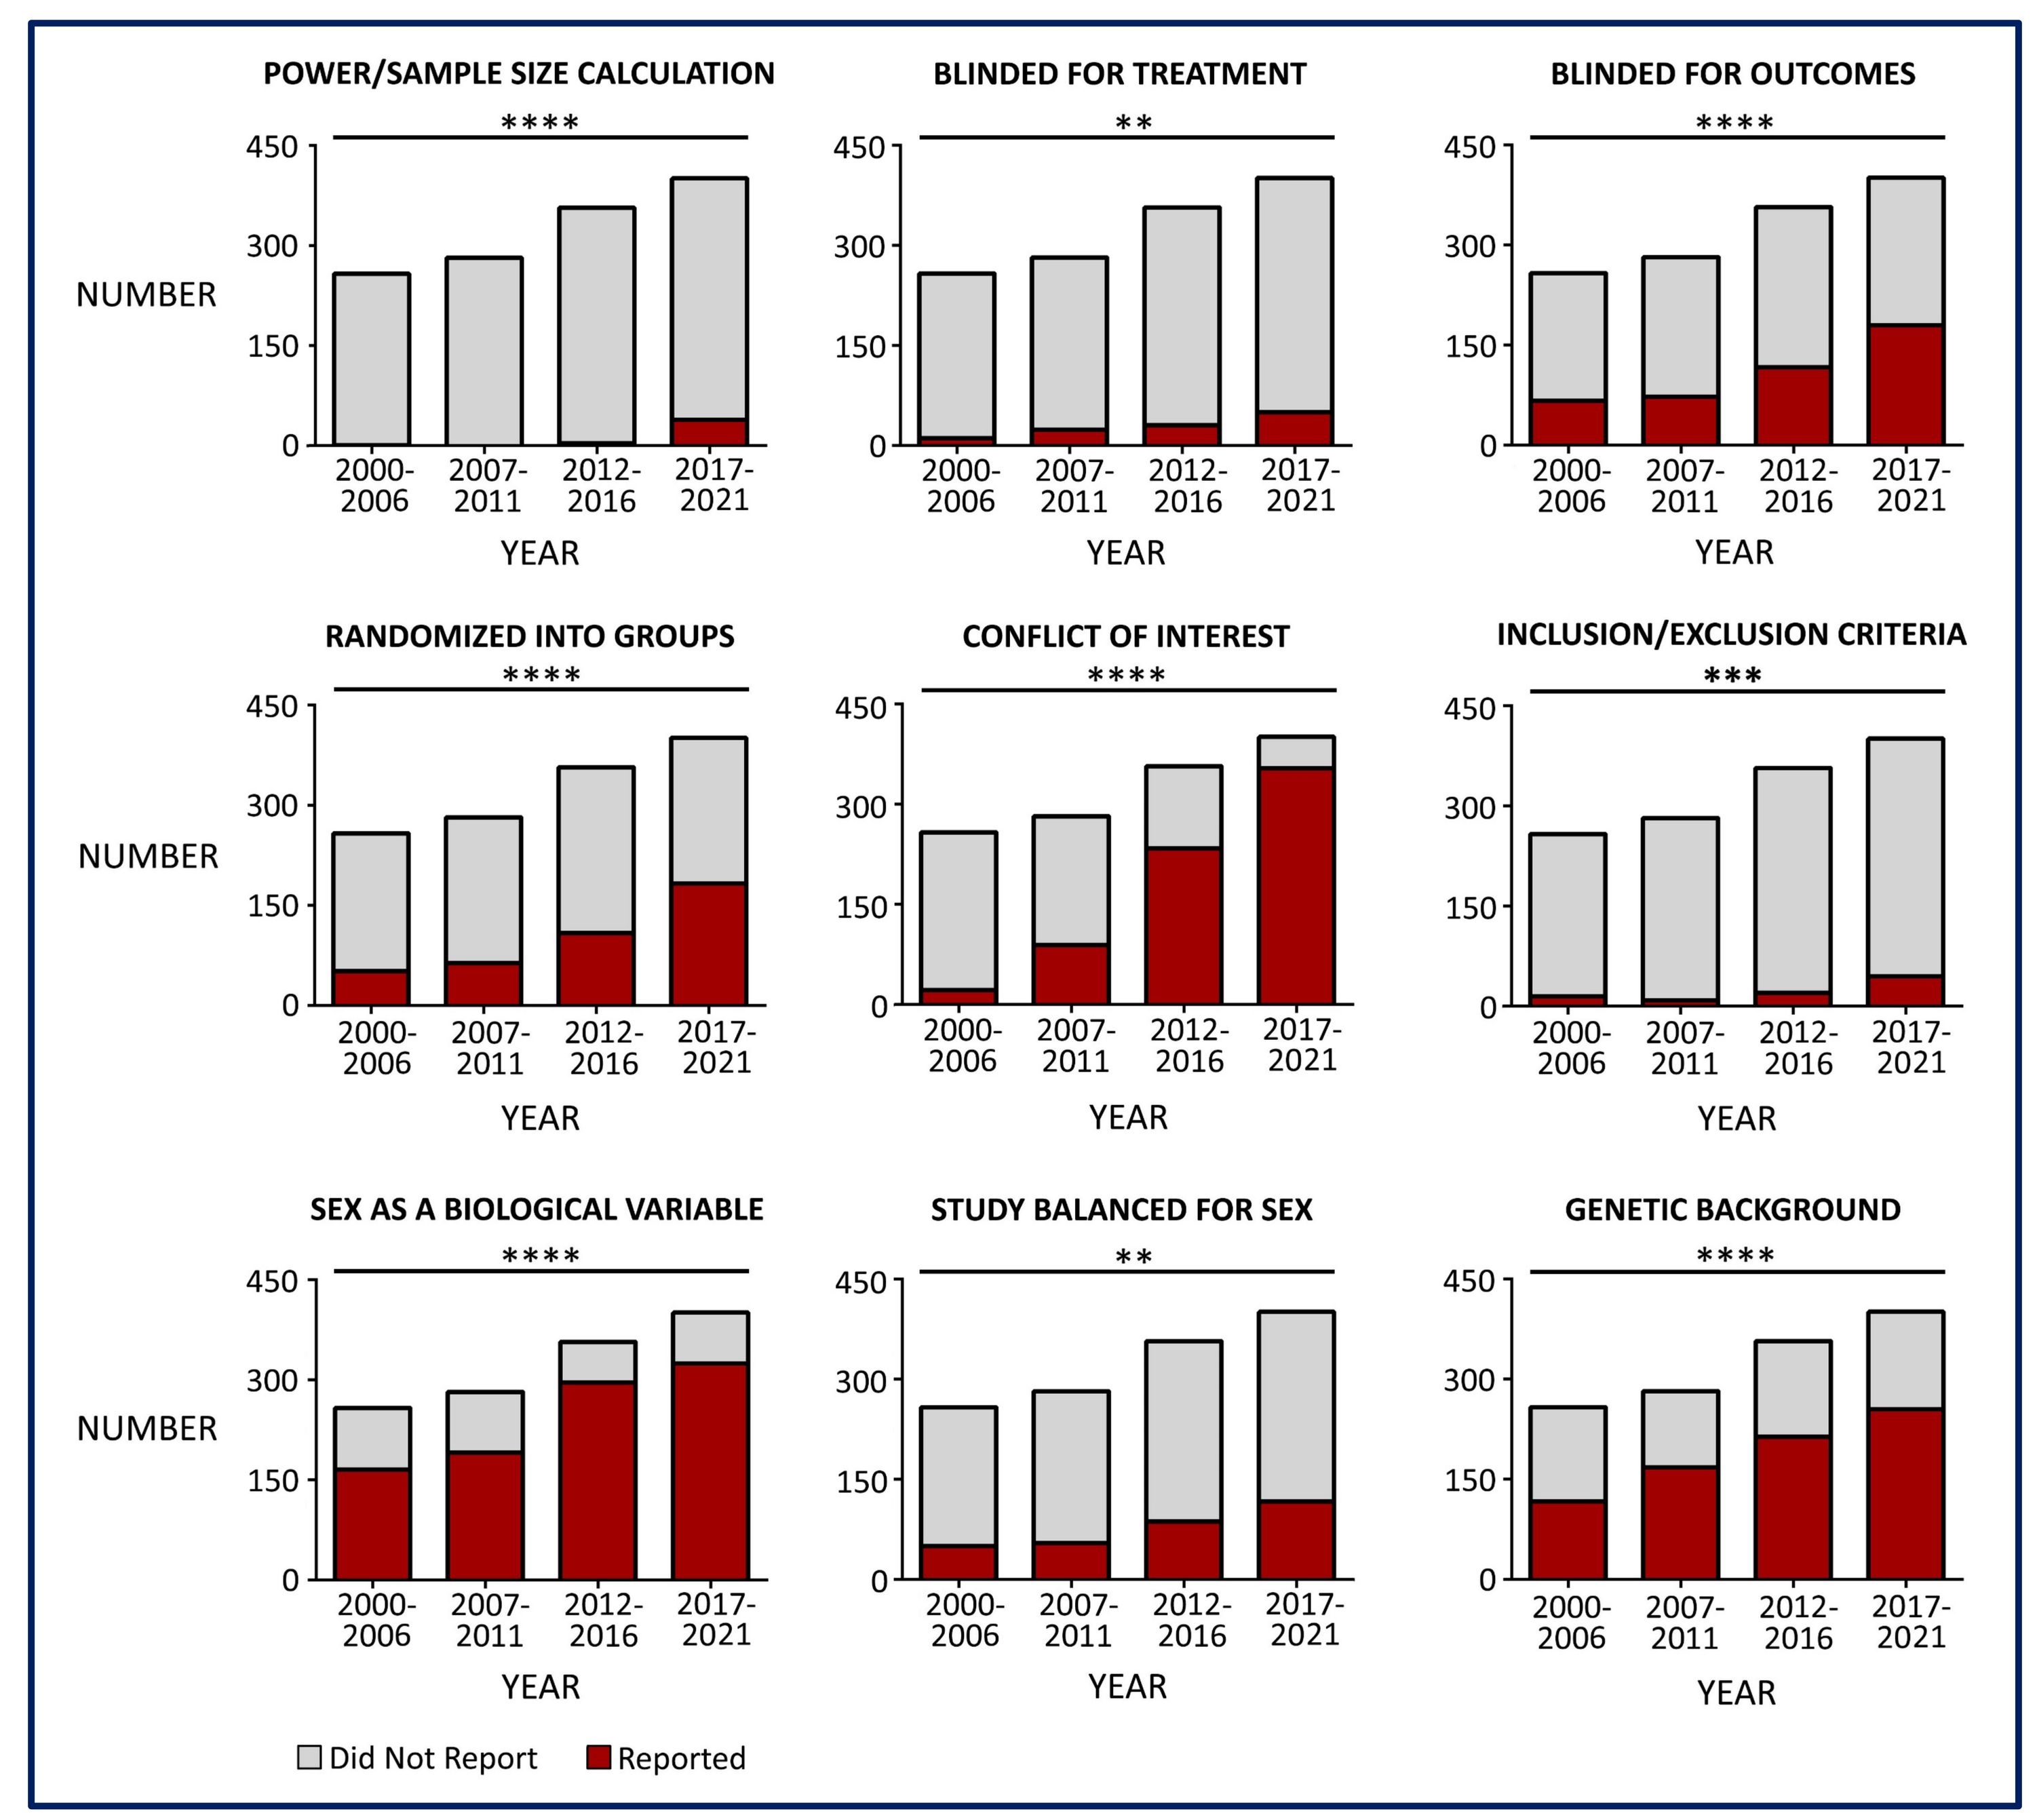

POWER/SAMPLE SIZE CALCULATION ****
BLINDED FOR TREATMENT **
BLINDED FOR OUTCOMES ****
RANDOMIZED INTO GROUPS ****
CONFLICT OF INTEREST ****
INCLUSION/EXCLUSION CRITERIA ***
SEX AS A BIOLOGICAL VARIABLE ****
STUDY BALANCED FOR SEX **
GENETIC BACKGROUND ****
NUMBER
0 150 300 450
2000-2006 2007-2011 2012-2016 2017-2021
YEAR
Did Not Report
Reported

Top: The graph shows the percentage of studies reporting the 9 core experimental design elements critical for scientific rigor and reproducibility. There is significant under-reporting of critical elements of methodology such as power calculation, blinding for treatment as well as for outcomes, randomization, inclusion/exclusion criteria and balancing for sex. Data is presented as percentage reported, calculated from 1298 published preclinical studies curated to AlzPED. The 9 core experimental design elements are derived from Shineman et al., 2011, Landis et al., 2012, Snyder et al., 2016 and ARRIVE guidelines.

Right: Graphs show reporting trends in the 9 critical core experimental design elements evaluated over 5-year spans from 2000 to 2021. There is steady improvement in some critical design elements like author conflict of interest statement, genetic background and sex of the animal model used in the study. However, other critical elements like power calculation, blinding, inclusion/exclusion criteria, and balancing a study for sex are still quite under-reported. Data are analyzed using Chi square test; **p<0.01, ***p<0.001, ****p<0.0001. Data presented as number that reported Vs number that did not report core experimental design elements, calculated from 258, 282, 357 and 401 curated studies published between 2000-2006, 2007-2011, 2012-2016 and 2017-2021 respectively.

UNPUBLISHED STUDY SUBMISSION PLATFORM

Submit unpublished preclinical efficacy study, with negative results to the AlzPED Unpublished Data Submission Portal

↓

Submitted study reviewed and curated by NIA experts for Bibliography, Therapeutics, Animal Model, Experimental Design and Outcomes

↓

- **Curated summary will be hosted on AlzPED**
- **Preprint pdf will be hosted on the AD Knowledge Portal**
- **A DOI will be generated for the preprint and is citable in grant applications, CVs and paper.**

Overview of the submission process for unpublished data. The DOI provided is citable in grant applications, CVs and peer-reviewed publications.

CONCLUSIONS

- Analysis of nearly 1300 curated studies demonstrates serious deficiencies in reporting critical elements of study design and methodology which diminish the scientific rigor, reproducibility and predictive value of preclinical therapeutic studies done in AD animal models.
- Analysis of reporting trends in the 9 core experimental design elements demonstrates improvements in the use of best practices between 2000-2006 and 2017-2021.
- Adoption of a standardized set of best practices as exemplified by study design elements in the AlzPED Rigor Report Card is very likely to improve the predictive validity of preclinical studies done in AD animal models. This measure is likely to promote the effective translation of preclinical drug testing data to the clinic
- AlzPED serves as a platform for reporting unpublished negative findings to mitigate publication bias that favors reporting of positive findings.

REFERENCES

- Shineman et al., Accelerating drug discovery for Alzheimer's disease: best practices for preclinical animal studies. *Alzheimers Res Ther. 2011; 3(5): 28. PMID: 21943025*.
- Landis et al., A call for transparent reporting to optimize the predictive value of preclinical research. *Nature 2012 Oct 11;490(7419):187-91. PMID: 23060188*.
- Snyder et al., Guidelines to improve animal study design and reproducibility for Alzheimer's disease and related dementias: For funders and researcher. *Alzheimers Dement. 2016 Nov;12(11):1177-1185. PMID: 27836053*.
- The ARRIVE Guidelines : https://arriveguidelines.org/

alzped.nia.nih.gov alzped@nih.gov @Alzheimers_NIH AlzPED

Detailed Analytics Summary is available here: AlzPED Analytics